Lentiviral vector cargo gene silencing during manufacturing

Sherin Parokkaran Johny[1]; Maria Ababi[1]; Harry Jenkins[1], Weimin Valenti[2], David Chang[2], Keith Meaney[1], Qian Liu[1], Maria I Patrício[1], Matthew Tridgett[1]
[1]OXGENE, Oxford, UK; [2]WuXi Advanced Therapies, Philadelphia, USA.

Introduction

- Lentiviral vectors (LVV) are commonly used for cargo gene delivery in cell and gene therapy.
- Although constitutive cargo gene expression in the target cells is desirable, during production it can be problematic for two reasons:
 - 1) Vector production can be inhibited either through metabolic burden placed on production cells or by inhibition of viral processes[1];
 - 2) Membrane-displayed gene products can be presented on the LVV surface since its envelope is derived from the production cell membrane[2] (Figure 1), potentially impacting downstream processing, transduction efficiency, and immunogenicity.

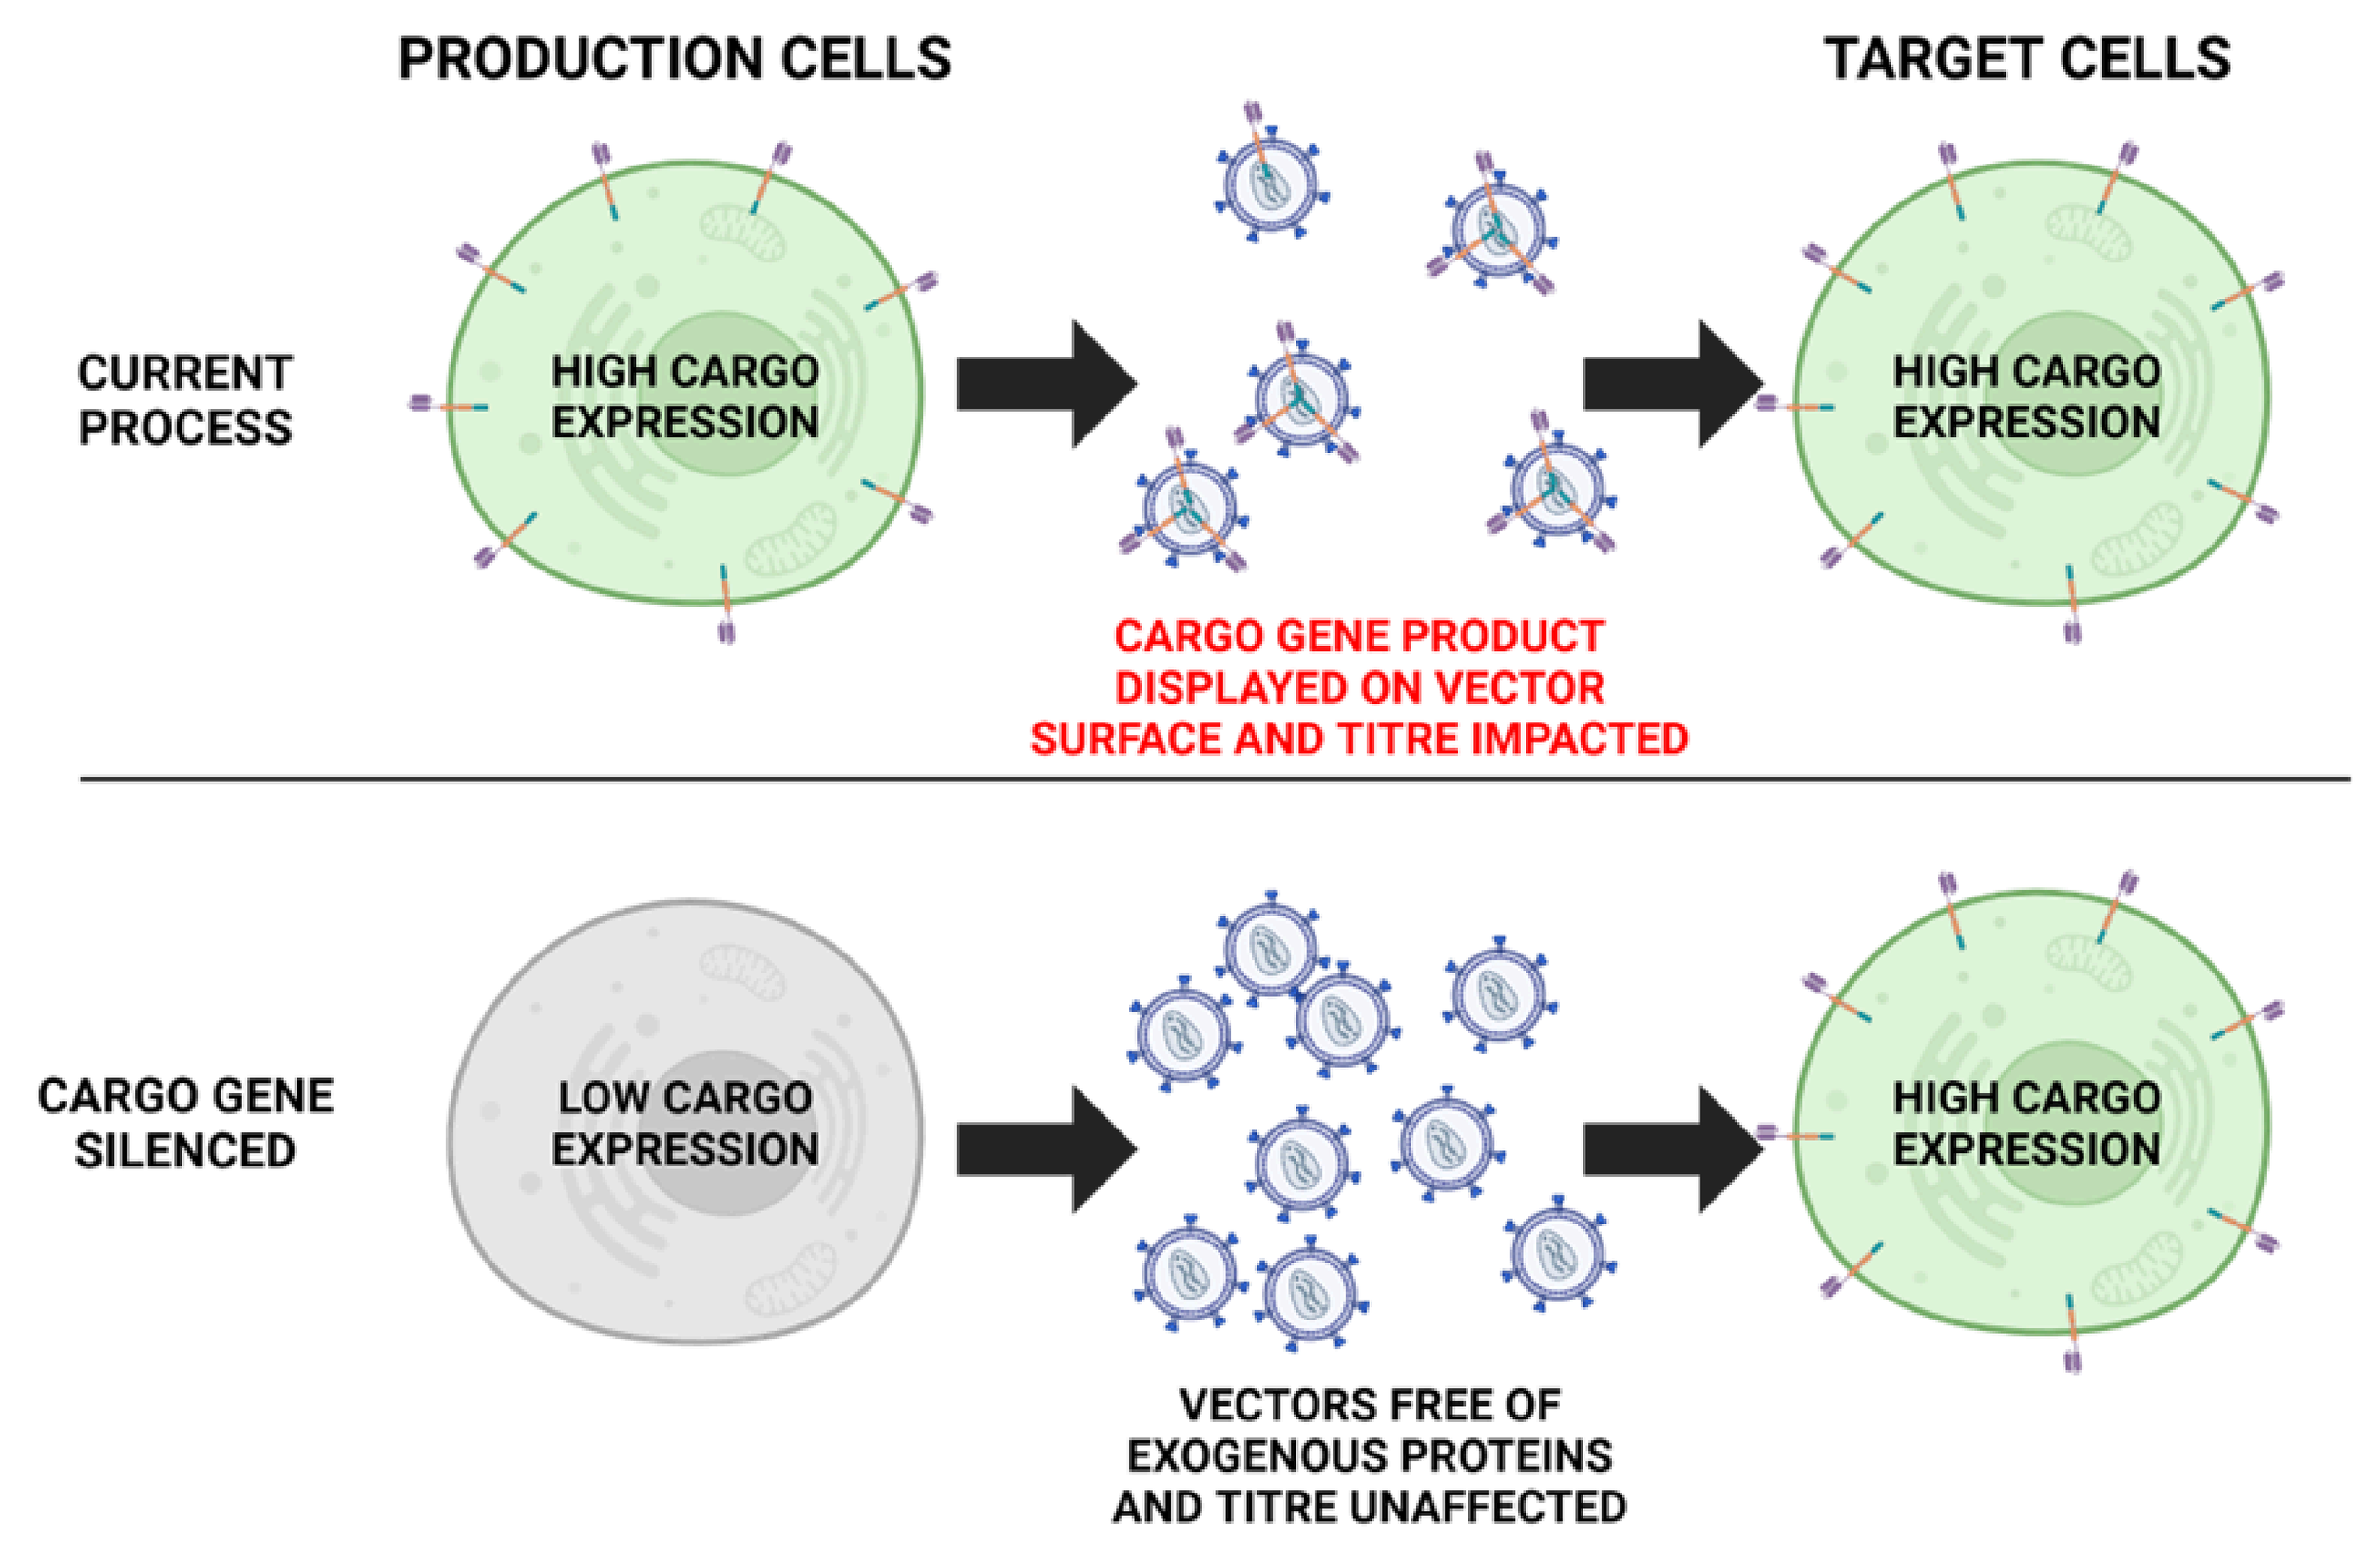
PRODUCTION CELLS
TARGET CELLS
CURRENT PROCESS
HIGH CARGO EXPRESSION
HIGH CARGO EXPRESSION
CARGO GENE PRODUCT DISPLAYED ON VECTOR SURFACE AND TITRE IMPACTED
CARGO GENE SILENCED
LOW CARGO EXPRESSION
HIGH CARGO EXPRESSION
VECTORS FREE OF EXOGENOUS PROTEINS AND TITRE UNAFFECTED

Figure 1. Advantages of cargo gene silencing during lentiviral vector manufacturing.

- An ideal production system would silence cargo gene expression and retain high titres. Thus, the viral RNA must not be damaged or inhibited. However, since the viral RNA contains the entire sequence of the cargo gene mRNA, typical silencing approaches would disrupt vector production. To address this challenge, we have engineered a distinction between the viral RNA and the cargo mRNA, enabling selective silencing of the cargo gene while leaving titres and cargo gene expression in target cells unaffected.
- Our system will enable the use of consistent upstream and downstream processes with different cargo genes, thus expediting, derisking, and reducing the cost of therapeutic development.

Cargo gene silencing challenge

- The cargo gene is encoded within the vector RNA, which is essential for vector transduction (Figure 2).

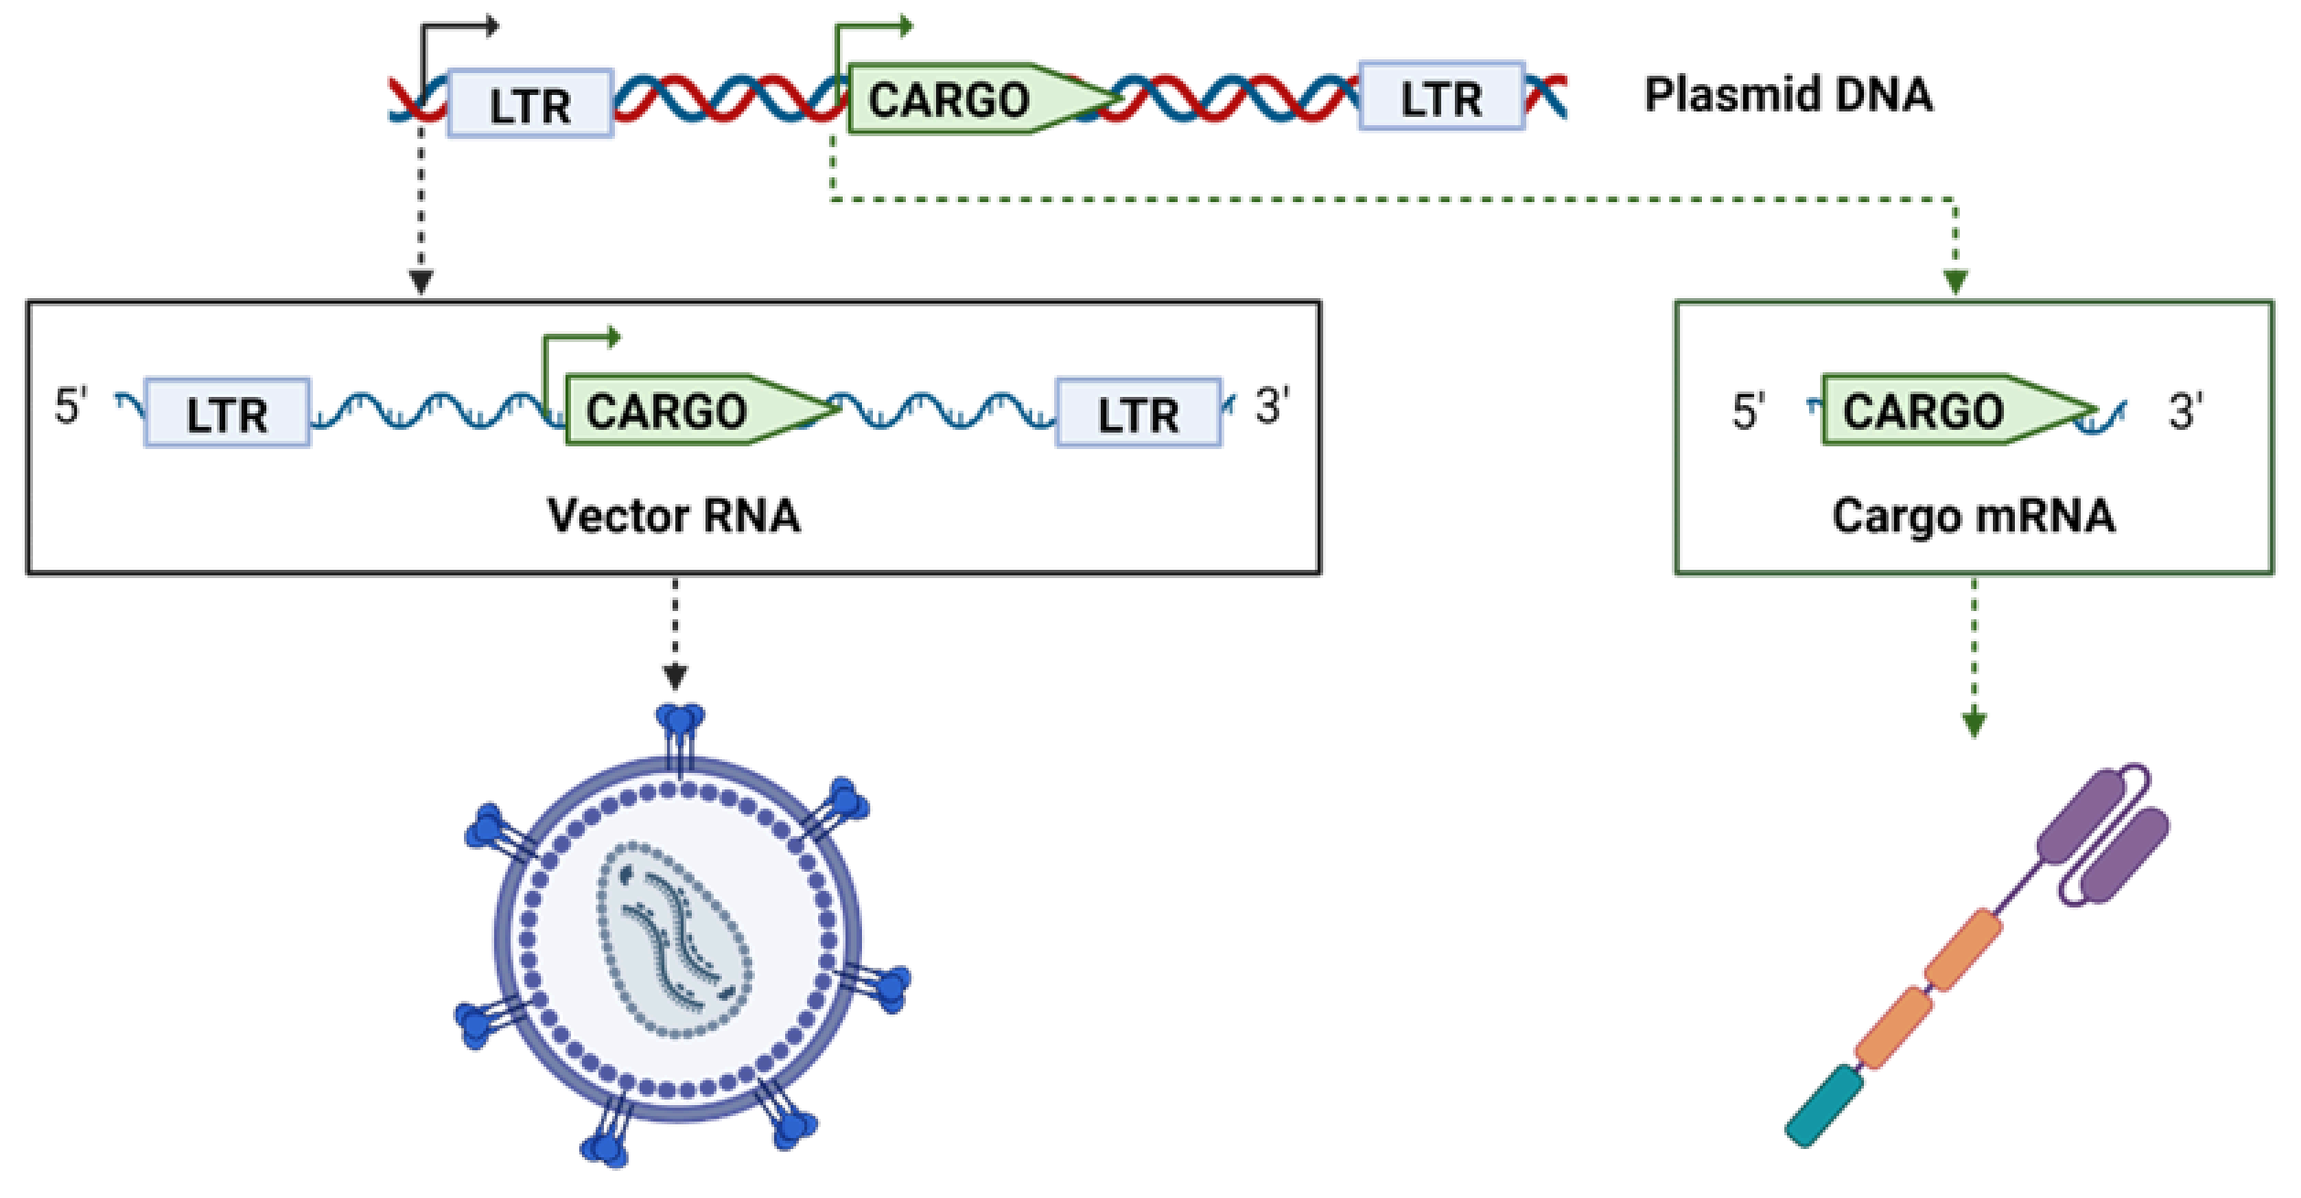
LTR
CARGO
LTR
Plasmid DNA
5' LTR CARGO LTR 3'
Vector RNA
5' CARGO 3'
Cargo mRNA

Figure 2. Lentiviral vector genome and transcripts.

- Thus, silencing the cargo gene by conventional means would be expected to negatively impact LVV titre (Figure 3).

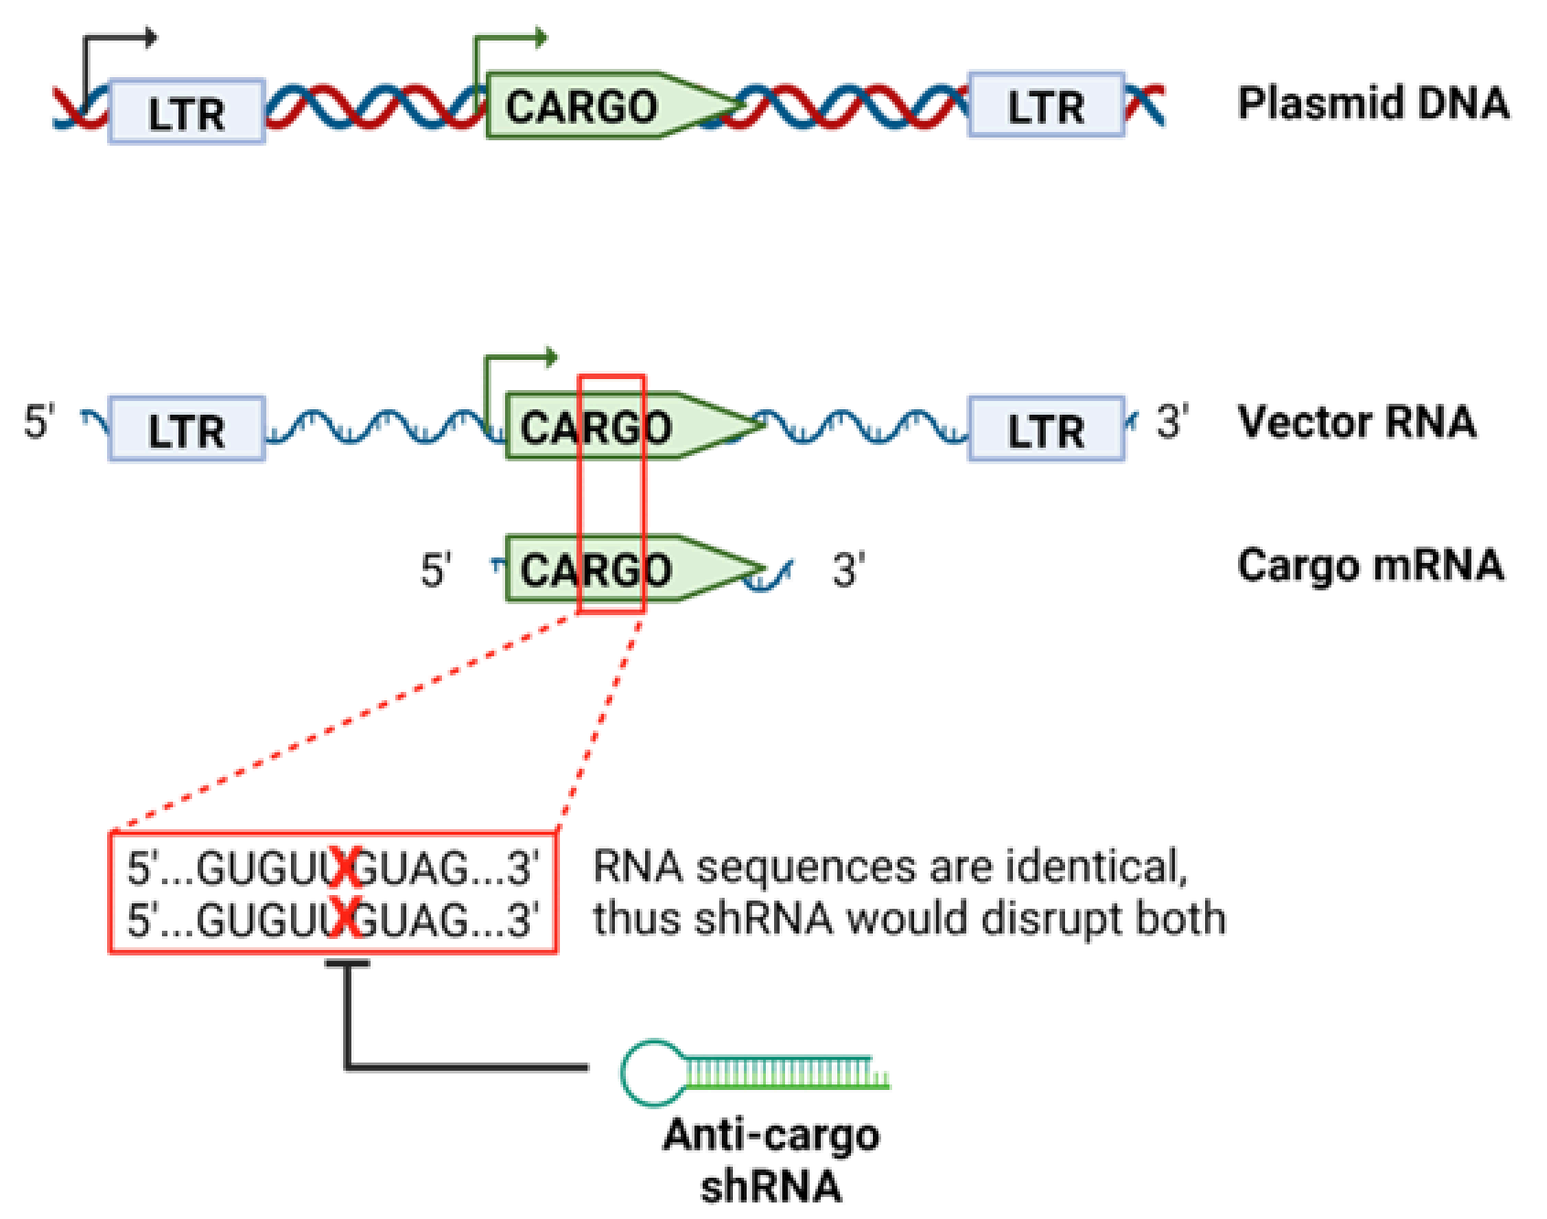
LTR
CARGO
LTR
Plasmid DNA
5' LTR CARGO LTR 3'
Vector RNA
5' CARGO 3'
Cargo mRNA
5'...GUGUUGUAG...3'
5'...GUGUUGUAG...3'
RNA sequences are identical, thus shRNA would disrupt both
Anti-cargo shRNA

Figure 3. Cargo gene silencing challenge.

- It is therefore necessary to engineer a distinction between the vector RNA and the cargo mRNA to enable selective cargo silencing without disruption to vector production.
- We describe here two iterations of development of an LVV silencing system: first, a two-plasmid silencing system; and second, a one-plasmid system.

Results (I)

Cargo gene silencing by two-plasmid system

- We first developed a two-plasmid silencing system (Figure 4 A). To make the system independent of cargo gene identity, we inserted a universal target site outside of the CDS (Figure 4 A). Thus, customisation requires only a single cloning step.
- To further improve the system, the universal target site chosen was a 21-bp fragment of an antiviral gene. Thus, the shRNA that silences the cargo gene also reduces antiviral gene expression during production.
- During LVV production, using the current process, we observed a large accumulation of cargo gene product (Figure 4 B).
- However, using the silencing system, we observed sustained cargo gene silencing, with up to 95% reduction in cargo gene expression (EGFP) compared to the current process (Figure 4 B).

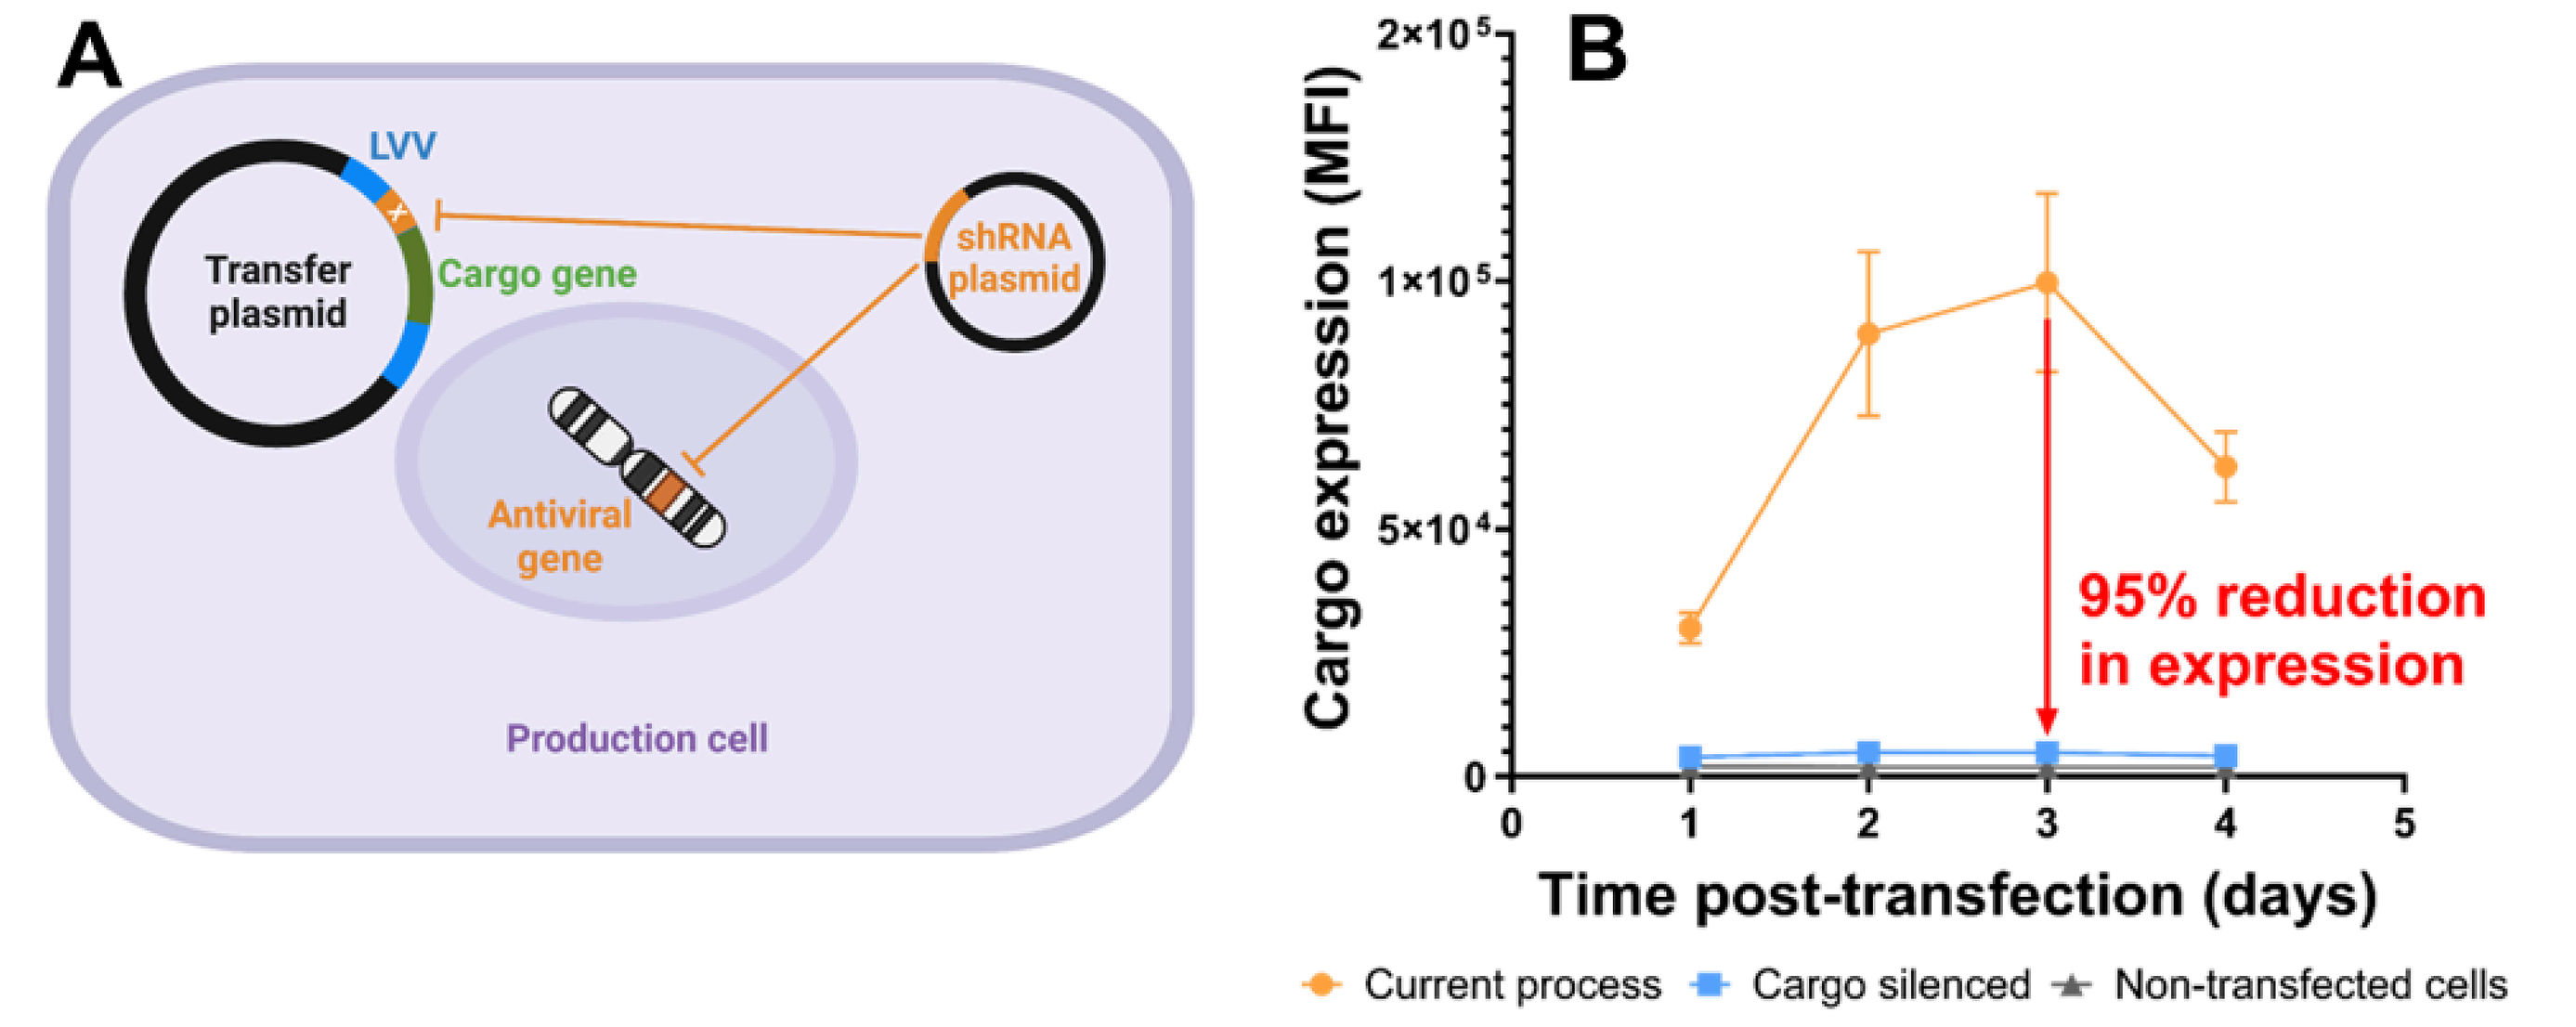
A
LVV
Transfer plasmid
Cargo gene
shRNA plasmid
Antiviral gene
Production cell
B
Cargo expression (MFI)
2×10⁵
1×10⁵
5×10⁴
0
95% reduction in expression
Time post-transfection (days)
Current process
Cargo silenced
Non-transfected cells

Figure 4. (**A**) Two-plasmid cargo gene silencing system. (**B**) Cargo gene expression during LVV production. N = 3 production replicates. Error bars indicate one standard deviation.

Experimental methods

- LVV productions were performed in 24 deep-well plate format, using the XOFLX™ Packaging cell line (requires transfection of cargo gene plasmid(s) only).
- Median fluorescence intensity (MFI) was measured by flow cytometry throughout production to monitor cargo gene (EGFP) silencing.
- Relative cargo gene expression = MFI of current process/Silenced MFI.
- All productions were performed in triplicate.

Results (II)

Cargo gene silencing by one-plasmid system

- We next developed a one-plasmid silencing system by inserting the shRNA cassette into the transfer plasmid (Figure 5 A).
- We tested the one-plasmid system with the cargo gene under control of the SFFV, EFS, or EF-1α promoters.
- With all cargo gene promoters, we observed sustained cargo gene silencing, with up to 98% reduction in cargo gene expression (EGFP) compared to the current process (Figure 5 B).

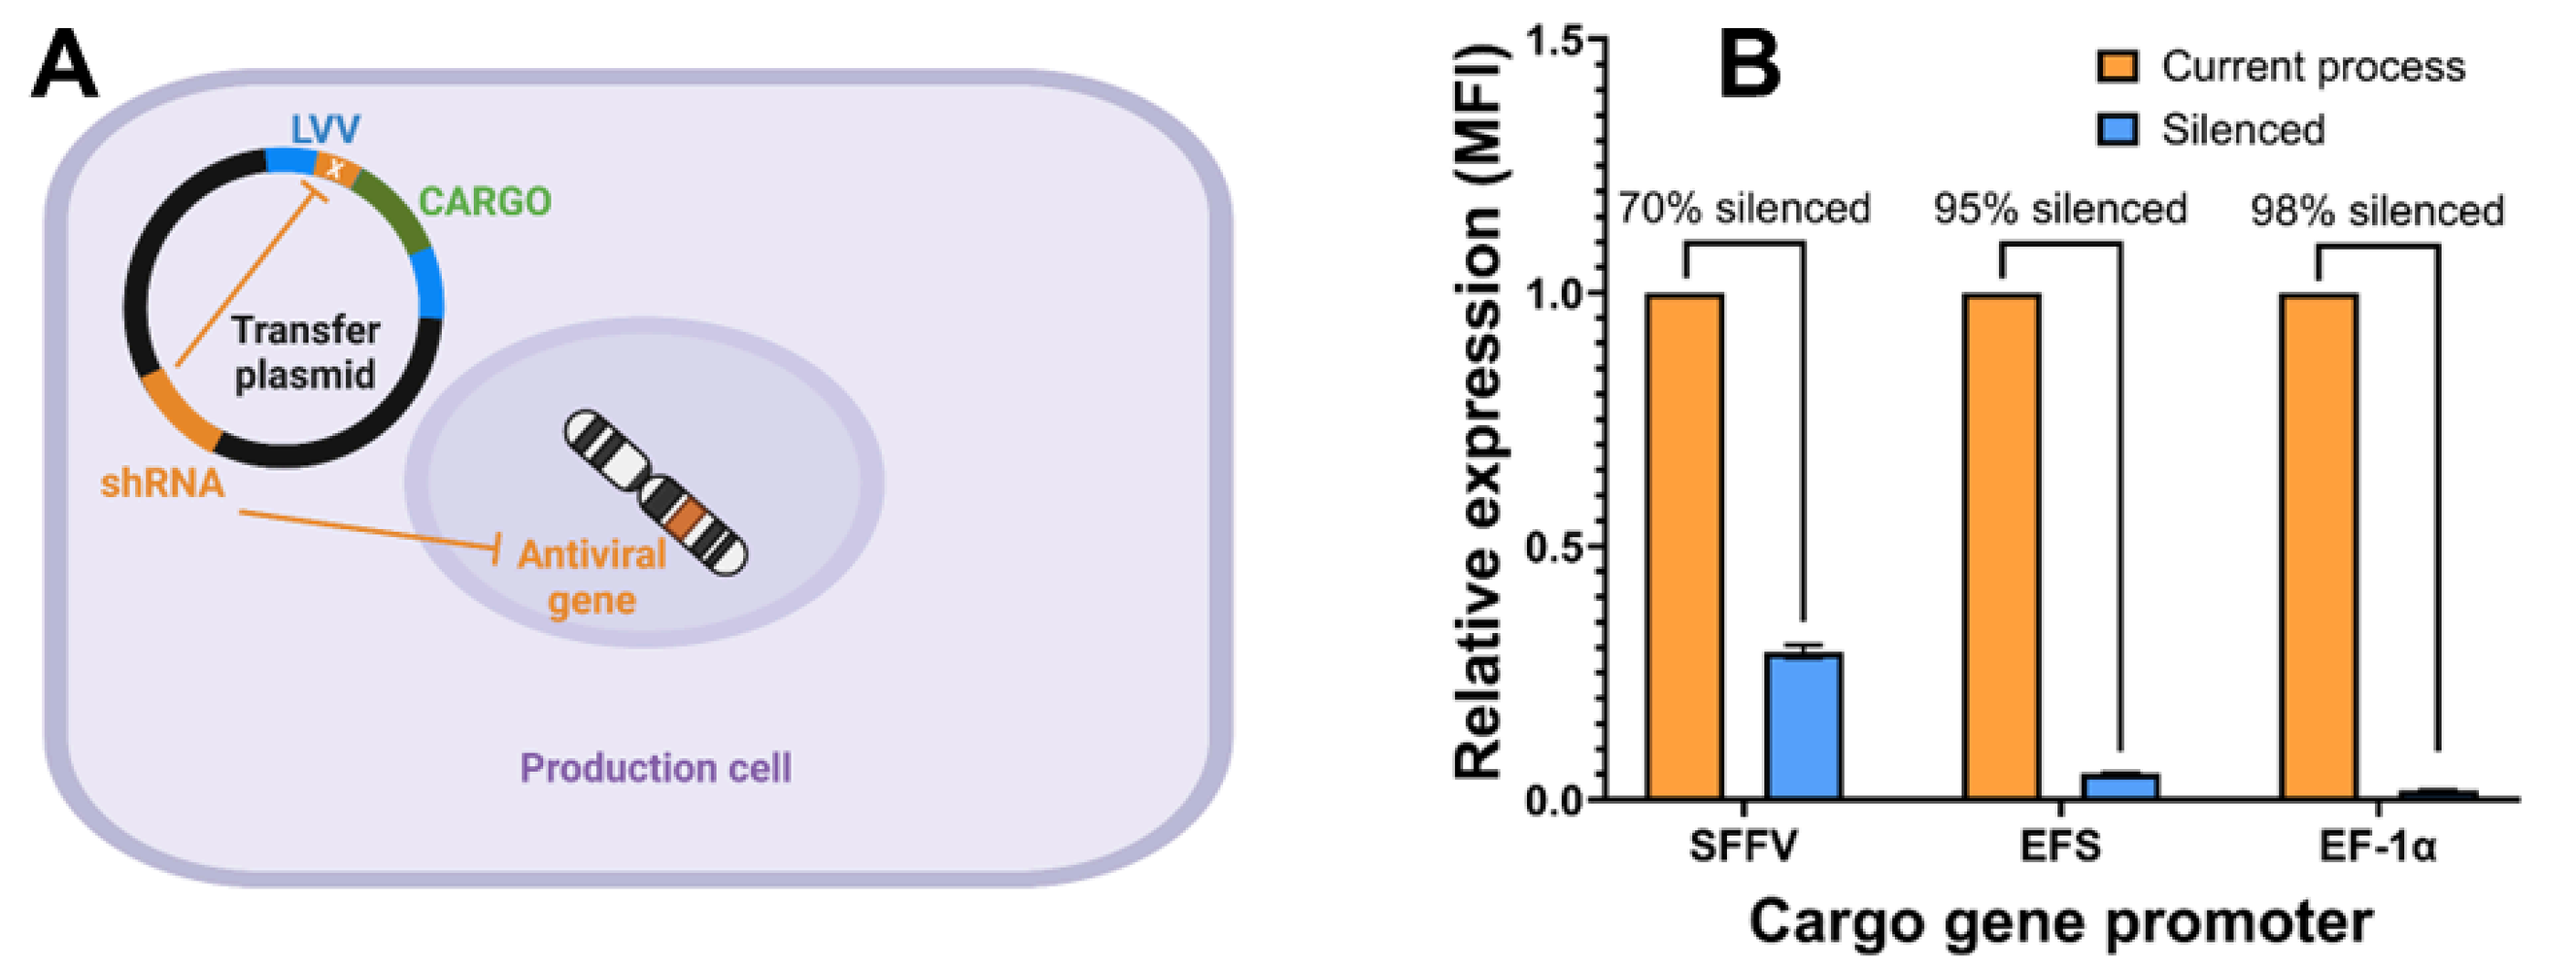
A
LVV
CARGO
Transfer plasmid
shRNA
Antiviral gene
Production cell
B
Current process
Silenced
Relative expression (MFI)
70% silenced
95% silenced
98% silenced
SFFV
EFS
EF-1α
Cargo gene promoter

Figure 5. (**A**) One-plasmid cargo gene silencing system. (**B**) Relative cargo gene expression with various promoters. N = 3 production replicates. Error bars indicate one standard deviation.

- Titration will be performed to test the selectivity of the silencing system.
- We expect that if a toxic cargo gene were encoded by the transfer plasmid, the silencing system would improve titres compared to the current process. This will form the basis of future experiments.

Conclusions

- Using our system, cargo gene expression has been reduced by up to 98% during lentiviral vector production.
- By engineering a distinction between the cargo mRNA and the viral RNA, we have overcome the challenges associated with silencing the cargo.
- We anticipate that with therapeutically relevant cargo genes, e.g. CARs, this technology will improve titres and particle quality, while derisking therapeutic development by reducing the number of unknowns associated with novel cargo genes.

Contact

4701 League Island Blvd, Philadelphia, PA 19112 | advancedtherapies.com
Corresponding author: mtridgett@oxgene.com | For information about our technologies, please contact us at business@oxgene.com

References

1. Radcliffe, P. A., Sion, C. J. M., Wilkes, F. J., Custard, E. J., Beard, G. L., Kingsman, S. M., and Mitrophanous, K. A. (2008). Analysis of factor VIII mediated suppression of lentiviral vector titres. *Gene Therapy*, 15(4), 289-297.
2. Chandrashekran, A., Gordon, M. Y., and Casimir, C. (2004). Targeted retroviral transduction of c-kit+ hematopoietic cells using novel ligand display technology. *Blood*, 104(9), 2697-2703.